A7475

CARDIAC EFFORT IS LESS VARIABLE THAN SIX-MINUTE WALK DISTANCE AND TRACKS WITH REVEAL LITE 2 RISK SCORE IN A SUBSTUDY OF THE PHASE 2 TORREY TRIAL OF SERALUTINIB IN PAH

Daniel J. Lachant[1], Robin Osterhout[2], Murali M. Chakinala[3], Veronica Franco[4], John W. McConnell[5], Franck F. Rahaghi[6], Sandeep Sahay[7], Namita Sood[8], Leslie A. Spikes[9], Raymond L. Benza[10], Richard N. Channick[11], Kelly M. Chin[12], Robert P. Frantz[13], Hossein-Ardeschir Ghofrani[14], Anna R. Hemnes[15], Luke S. Howard[16], Vallerie V. McLaughlin[17], Olivier Sitbon[18], Roham T. Zamanian[19], Robert F. Roscigno[2], David Mottola[2], Ed Parsley[2], Matt Cravets[2], Jean-Marie Bruey[2], Lawrence S. Zisman[2], R. James White[20]

[1]University of Rochester Medical Center, Rochester, NY, USA; [2]Gossamer Bio, Inc., San Diego, CA, USA; [3]Washington University School of Medicine, St Louis, MO, USA; [4]The Ohio State University Wexner Medical Center, Columbus, OH, USA; [5]Norton Healthcare, Louisville, KY, USA; [6]Cleveland Clinic Florida, Weston, FL, USA; [7]Houston Methodist Hospital/Weill Cornell Medicine, Houston, TX, USA; [8]University of California Davis Medical Center, Sacramento, CA, USA; [9]University of Kansas Medical Center, Kansas City, KS, USA; [10]Icahn School of Medicine at Mount Sinai, Mount Sinai Hospital, New York, NY, USA; [11]UCLA Medical Center, Los Angeles, CA, USA; [12]UT Southwestern Medical Center, Dallas, TX, USA; [13]Mayo Clinic, Rochester, MN, USA; [14]Justus-Liebig-University Giessen and Marburg Lung Center (UGMLC), Institute for Lung Health, Cardio-Pulmonary Institute; Member of the German Center for Lung Research (DZL), Giessen, Germany; Department of Medicine, Imperial College, London, UK; [15]Vanderbilt University, Nashville, TN, USA; [16]Hammersmith Hospital, Imperial College Healthcare NHS Trust, London, UK; [17]University of Michigan, Ann Arbor, MI, USA; [18]Hôpital Bicêtre (AP-HP), Université Paris-Saclay, Le Kremlin-Bicêtre, France; [19]Stanford University School of Medicine, Stanford, CA, USA; [20]Division of Pulmonary and Critical Care Medicine, University of Rochester Medical Center, Rochester, NY, USA

TORREY STUDY

Presented at the ATS International Conference
San Diego, CA, USA
May 17-22, 2024

BACKGROUND

- **6-Minute Walk Distance (6MWD)** is a core endpoint in pulmonary arterial hypertension (PAH) clinical trials and heavily weighted in PAH risk assessments. Intrinsic variability for longer 6MWD makes interpreting changes difficult for more functional patients[1,2]
- **Cardiac Effort (CE)**, the number of heartbeats used during 6-minute walk test (6MWT)/6MWD, correlates with stroke volume, is less variable than 6MWD, and decreases with clinical improvement[3-5]
- **Hypothesis:** Cardiac effort will
 1. have less variability in placebo-treated patients than 6MWD
 2. decrease with seralutinib
 3. correlate with clinical metrics (eg, right ventricular function, risk)

METHODS

- This exploratory CE substudy was conducted as part of the phase 2 TORREY study of seralutinib in adults with PAH (NCT04456998)[6]
- Continuous heart rate monitoring with an electrocardiogram (ECG) monitor (VivaLink patch, worn on chest) was performed during 6MWT at Baseline, Week 12, and Week 24. Heart rate data were transmitted from the sensor to an app through Bluetooth and uploaded to a secure server. LabChart 8 (ADInstrument) was used to analyze ECG data. Heart rates were averaged every 4 seconds, and CE was calculated as total number of heartbeats during 6MWT/6MWD
- Due to the pandemic, site instruction was given remotely. In several instances, data were lost due to transmission failures and, consequently, not all consenting patients had data for all 3 walks. This poster focuses on those patients with Baseline and follow-up 6MWTs (n=14)
- Friedman test of repeated measures was performed to evaluate changes in CE between Baseline, Week 12, and Week 24 in seralutinib- and placebo-treated patients. One seralutinib-assigned patient had a 6MWT at Week 24 with 6MWD similar to Week 12; but VivaLink data at Week 24 were lost. For this patient, we imputed the total number of heartbeats at Week 24 as last observation carried forward (LOCF). Non-parametric testing with Mann-Whitney test was used to evaluate changes from Baseline between treatment groups. Pearson correlations were performed with normally distributed data. Spearman correlations were performed for non-normally distributed data

RESULTS

Table. Baseline demographics and clinical variables in seralutinib- and placebo-treated patients. Data are summarized as median (interquartile range [IQR]) unless otherwise noted. Several Baseline imbalances in this substudy suggest that seralutinib-assigned patients had more severe disease.

Characteristic	All patients in CE substudy		Patients with repeated CE measures	
	Placebo (n=11)	Seralutinib (n=9)	Placebo (n=7)	Seralutinib (n=7)
Age, y	45 (40, 56)	51 (40, 57)	48 (40, 64)	54 (45, 57)
Female, n (%)	11 (100)	8 (89)	7 (100)	6 (86)
Type of PAH, n (%)				
Idiopathic/Heritable	8 (73)	6 (67)	4 (57)	5 (72)
Associated CTD	3 (27)	1 (11)	3 (43)	1 (14)
Associated anorexia/meth	0	2 (22)	0	1 (14)
PAH therapy, n (%)				
ERA + PDE5i or SGC	7 (64)	4 (45)	4 (57)	2 (29)
PDE5i + prostacyclin	1 (9)	0	0	0
Combo + prostacyclin	3 (27)	5 (55)	3 (43)	5 (71)
NT-proBNP, pg/mL	142 (41, 304)	881 (106, 1758)	215 (27, 462)	881 (152, 2088)
6MWD, m	452 (415, 485)	375 (314, 450)	426 (415, 457)	375 (322, 439)
Cardiac effort, beats/m	1.6 (1.5, 1.8)	1.8 (1.5, 1.9)	1.7 (1.5, 1.8)	1.8 (1.6, 2.0)
Peak heart rate, beats/min	137 (122, 144)	121 (100, 124)	128 (118, 137)	121 (106, 123)
Heart rate expenditure, beats	740 (656, 785)	658 (575, 691)	736 (655, 785)	673 (597, 694)
REVEAL Lite 2 risk score	3 (2, 4)	6 (4, 9)	4 (3, 4)	6 (5, 9)
Echocardiogram				
RVFAC, %	38.1 (32.1, 42.7)	28.4 (23.8, 40.9)	40.2 (32.0, 43.2)	30.4 (23.0, 41.6)
TAPSE, cm	1.54 (1.30, 1.85)	1.79 (1.55, 2.11)	1.58 (1.32, 1.99)	1.79 (1.6, 2.2)

6MWD, six-minute walk distance; CTD, connective tissue disease; ERA, endothelin receptor antagonist; meth, methamphetamine use; NT-proBNP, N-terminal pro-brain natriuretic peptide; PAH, pulmonary arterial hypertension; PDE5i, phosphodiesterase 5 inhibitor; SGC, soluble guanylate cyclase; TAPSE, tricuspid annular plane systolic excursion; REVEAL, Registry to Evaluate Early and Long-term PAH Disease Management; RVFAC, right ventricular fractional area change.

RESULTS *(continued)*

Median cardiac effort decreased and 6MWD increased following treatment with seralutinib; both were stable with placebo.

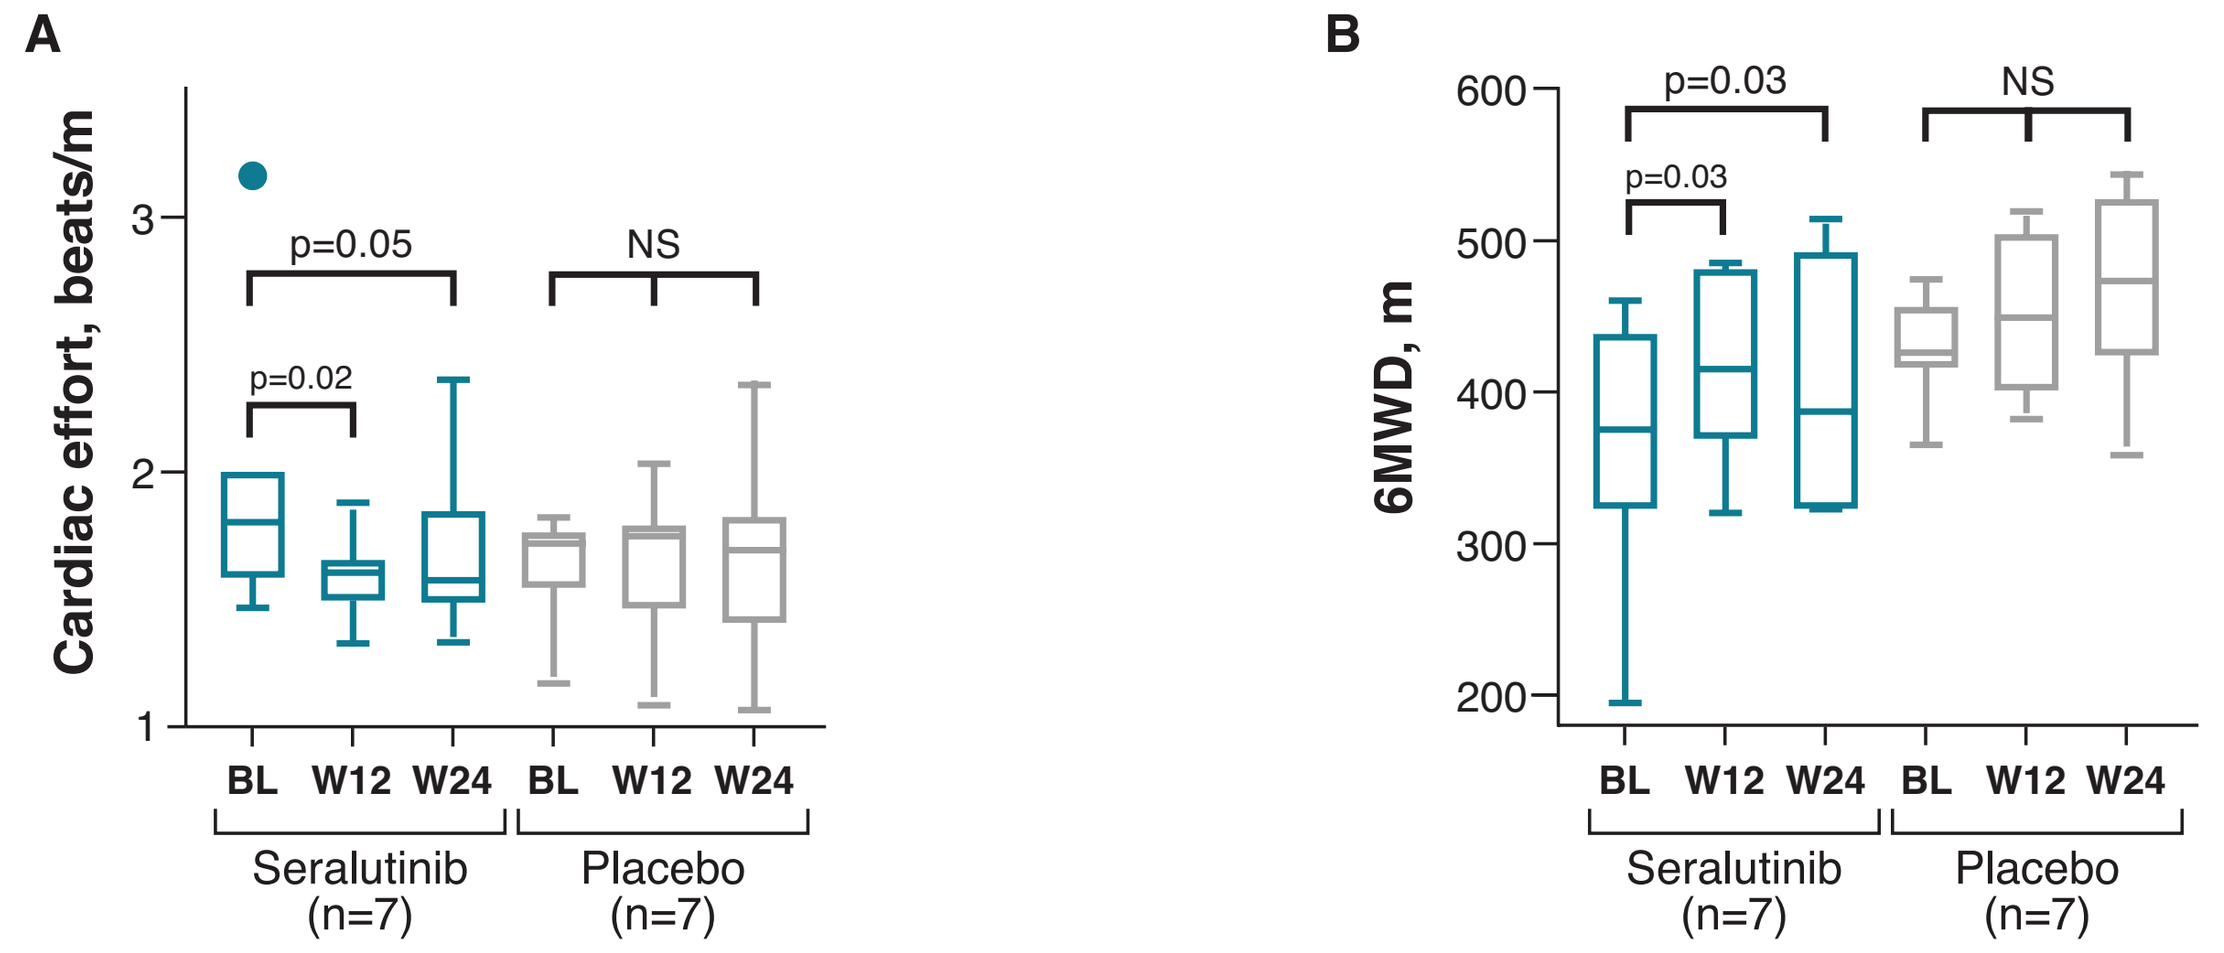
A
Cardiac effort, beats/m
p=0.05
p=0.02
NS
BL W12 W24 BL W12 W24
Seralutinib (n=7)
Placebo (n=7)
B
6MWD, m
p=0.03
p=0.03
NS
BL W12 W24 BL W12 W24
Seralutinib (n=7)
Placebo (n=7)

Figure 1. Boxplots of CE and 6MWD values at each visit. A. CE in seralutinib-treated patients decreased at Weeks 12 and 24 vs Baseline; no change in placebo-treated patients. One seralutinib-treated patient did not have Week 24 heart rate data; LOCF (Week 12) was used to impute heart rate for Week 24. **B.** An increase in 6MWD in seralutinib-treated patients was observed for the cohort at Weeks 12 and 24. No change in placebo. Friedman's repeated measures with post-hoc Dunn's corrected comparisons. 6MWD, 6-minute walk distance; BL, Baseline; CE, cardiac effort; LOCF, last observation carried forward; NS, not significant; W, week.

Cardiac effort decreased at Week 12 with seralutinib treatment relative to placebo; variability (IQR) of change in 6MWD was higher than variability of change in cardiac effort.

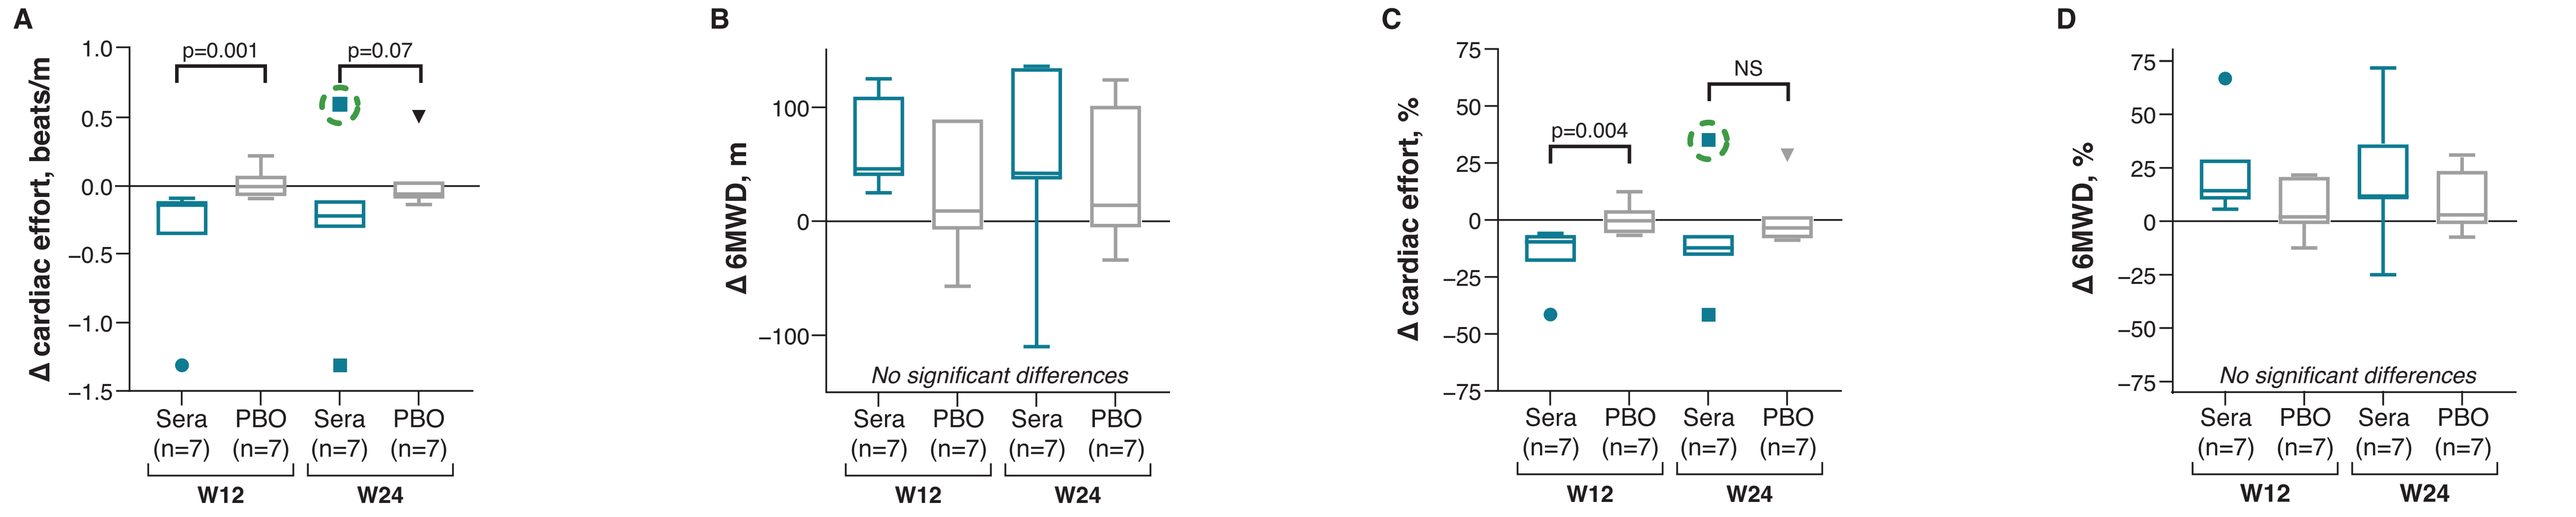
A
Δ cardiac effort, beats/m
p=0.001
p=0.07
B
Δ 6MWD, m
No significant differences
C
Δ cardiac effort, %
p=0.004
NS
D
Δ 6MWD, %
No significant differences
Sera (n=7) PBO (n=7) Sera (n=7) PBO (n=7)
W12 W24

Figure 2 A, B. Absolute change in CE from Baseline to Week 12 or 24 shows the stability of the CE measure in placebo patients compared to absolute change in 6MWD; there appears to be a shift from Baseline for CE in seralutinib-treated vs placebo-treated patients. **C**, **D**. 6MWD, expressed as % change, is much more variable in both treatment groups. Outlier seralutinib patient (circled in green) *was clinically worse at Week 24 by all measures* (CE accurately flagged her deterioration seen on echocardiogram and catheterization). In this small cohort, this patient had a large impact on the results. 6MWD, 6-minute walk distance; CE, cardiac effort; IQR, interquartile range; NS, not significant; PBO, placebo; Sera, seralutinib; W, week.

Change in cardiac effort had a stronger correlation with ΔNT-proBNP than Δ6MWD; reductions in cardiac effort are consistent with reduced (improved, green boxplot) REVEAL Lite 2 risk score.

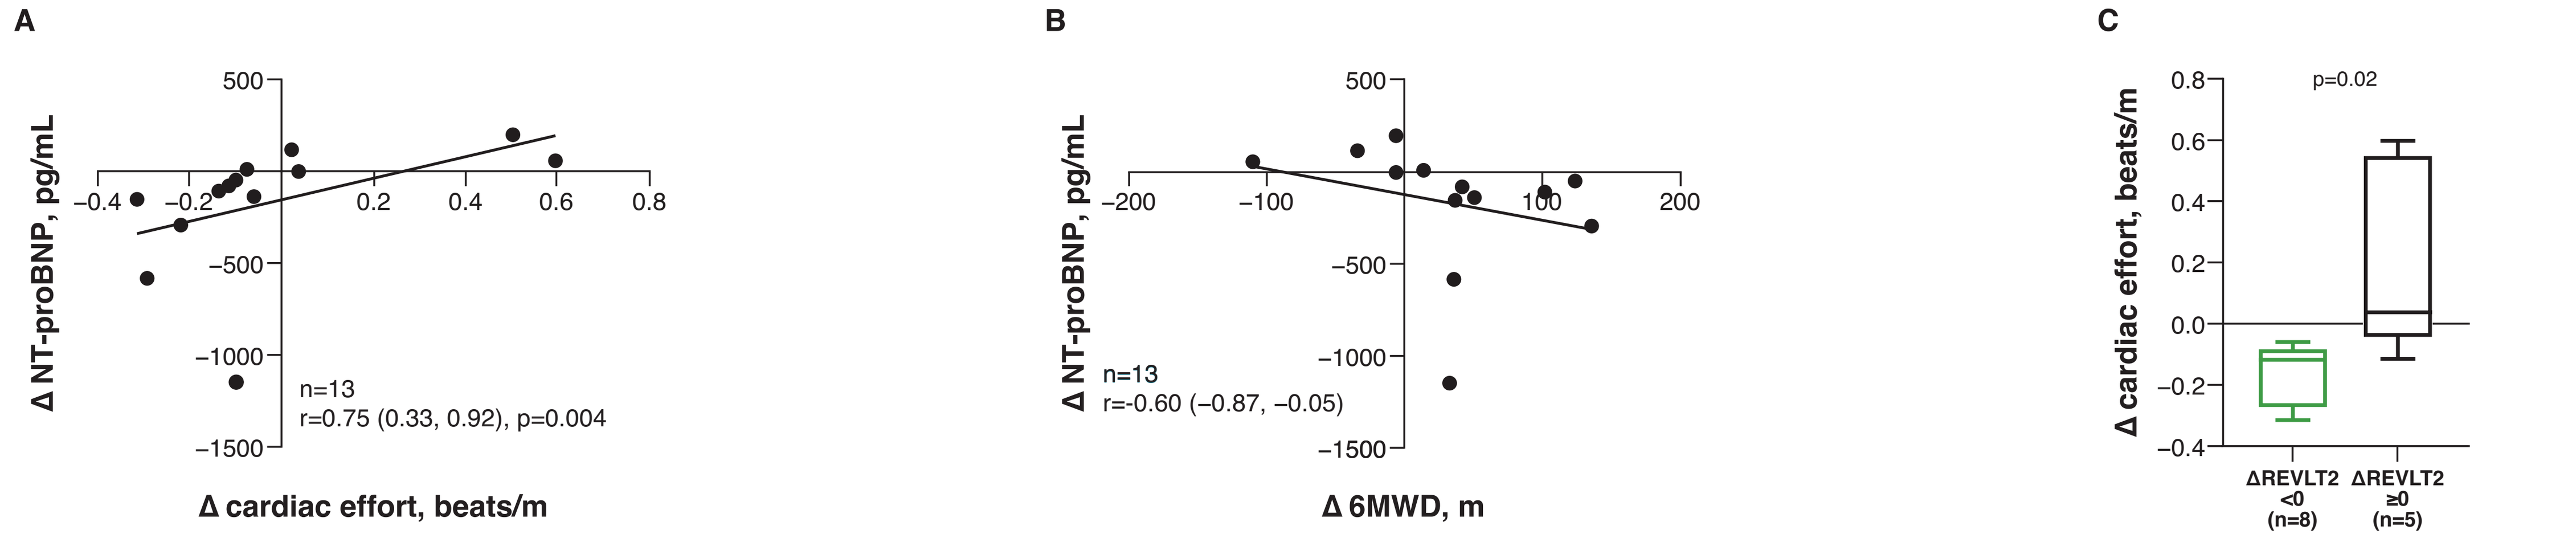
A
Δ NT-proBNP, pg/mL
n=13
r=0.75 (0.33, 0.92), p=0.004
Δ cardiac effort, beats/m
B
Δ NT-proBNP, pg/mL
n=13
r=-0.60 (−0.87, −0.05)
Δ 6MWD, m
C
Δ cardiac effort, beats/m
p=0.02
ΔREVLT2 <0 (n=8)
ΔREVLT2 ≥0 (n=5)

Figure 3. Spearman correlations between ΔNT-proBNP and **A.** ΔCE or **B.** Δ6MWD; R is higher with ΔCE and confidence intervals are tighter. **C.** CE changes are directionally consistent with REVEAL Lite 2 (ΔREVLT2) risk score changes. Graphs show pooled placebo and seralutinib data. 6MWD, 6-minute walk distance; CE, cardiac effort; NT-proBNP, N-terminal pro-brain natriuretic peptide; REVEAL, Registry to Evaluate Early and Long-term PAH Disease Management.

Cardiac effort showed higher correlations with echocardiogram parameters than 6MWD.

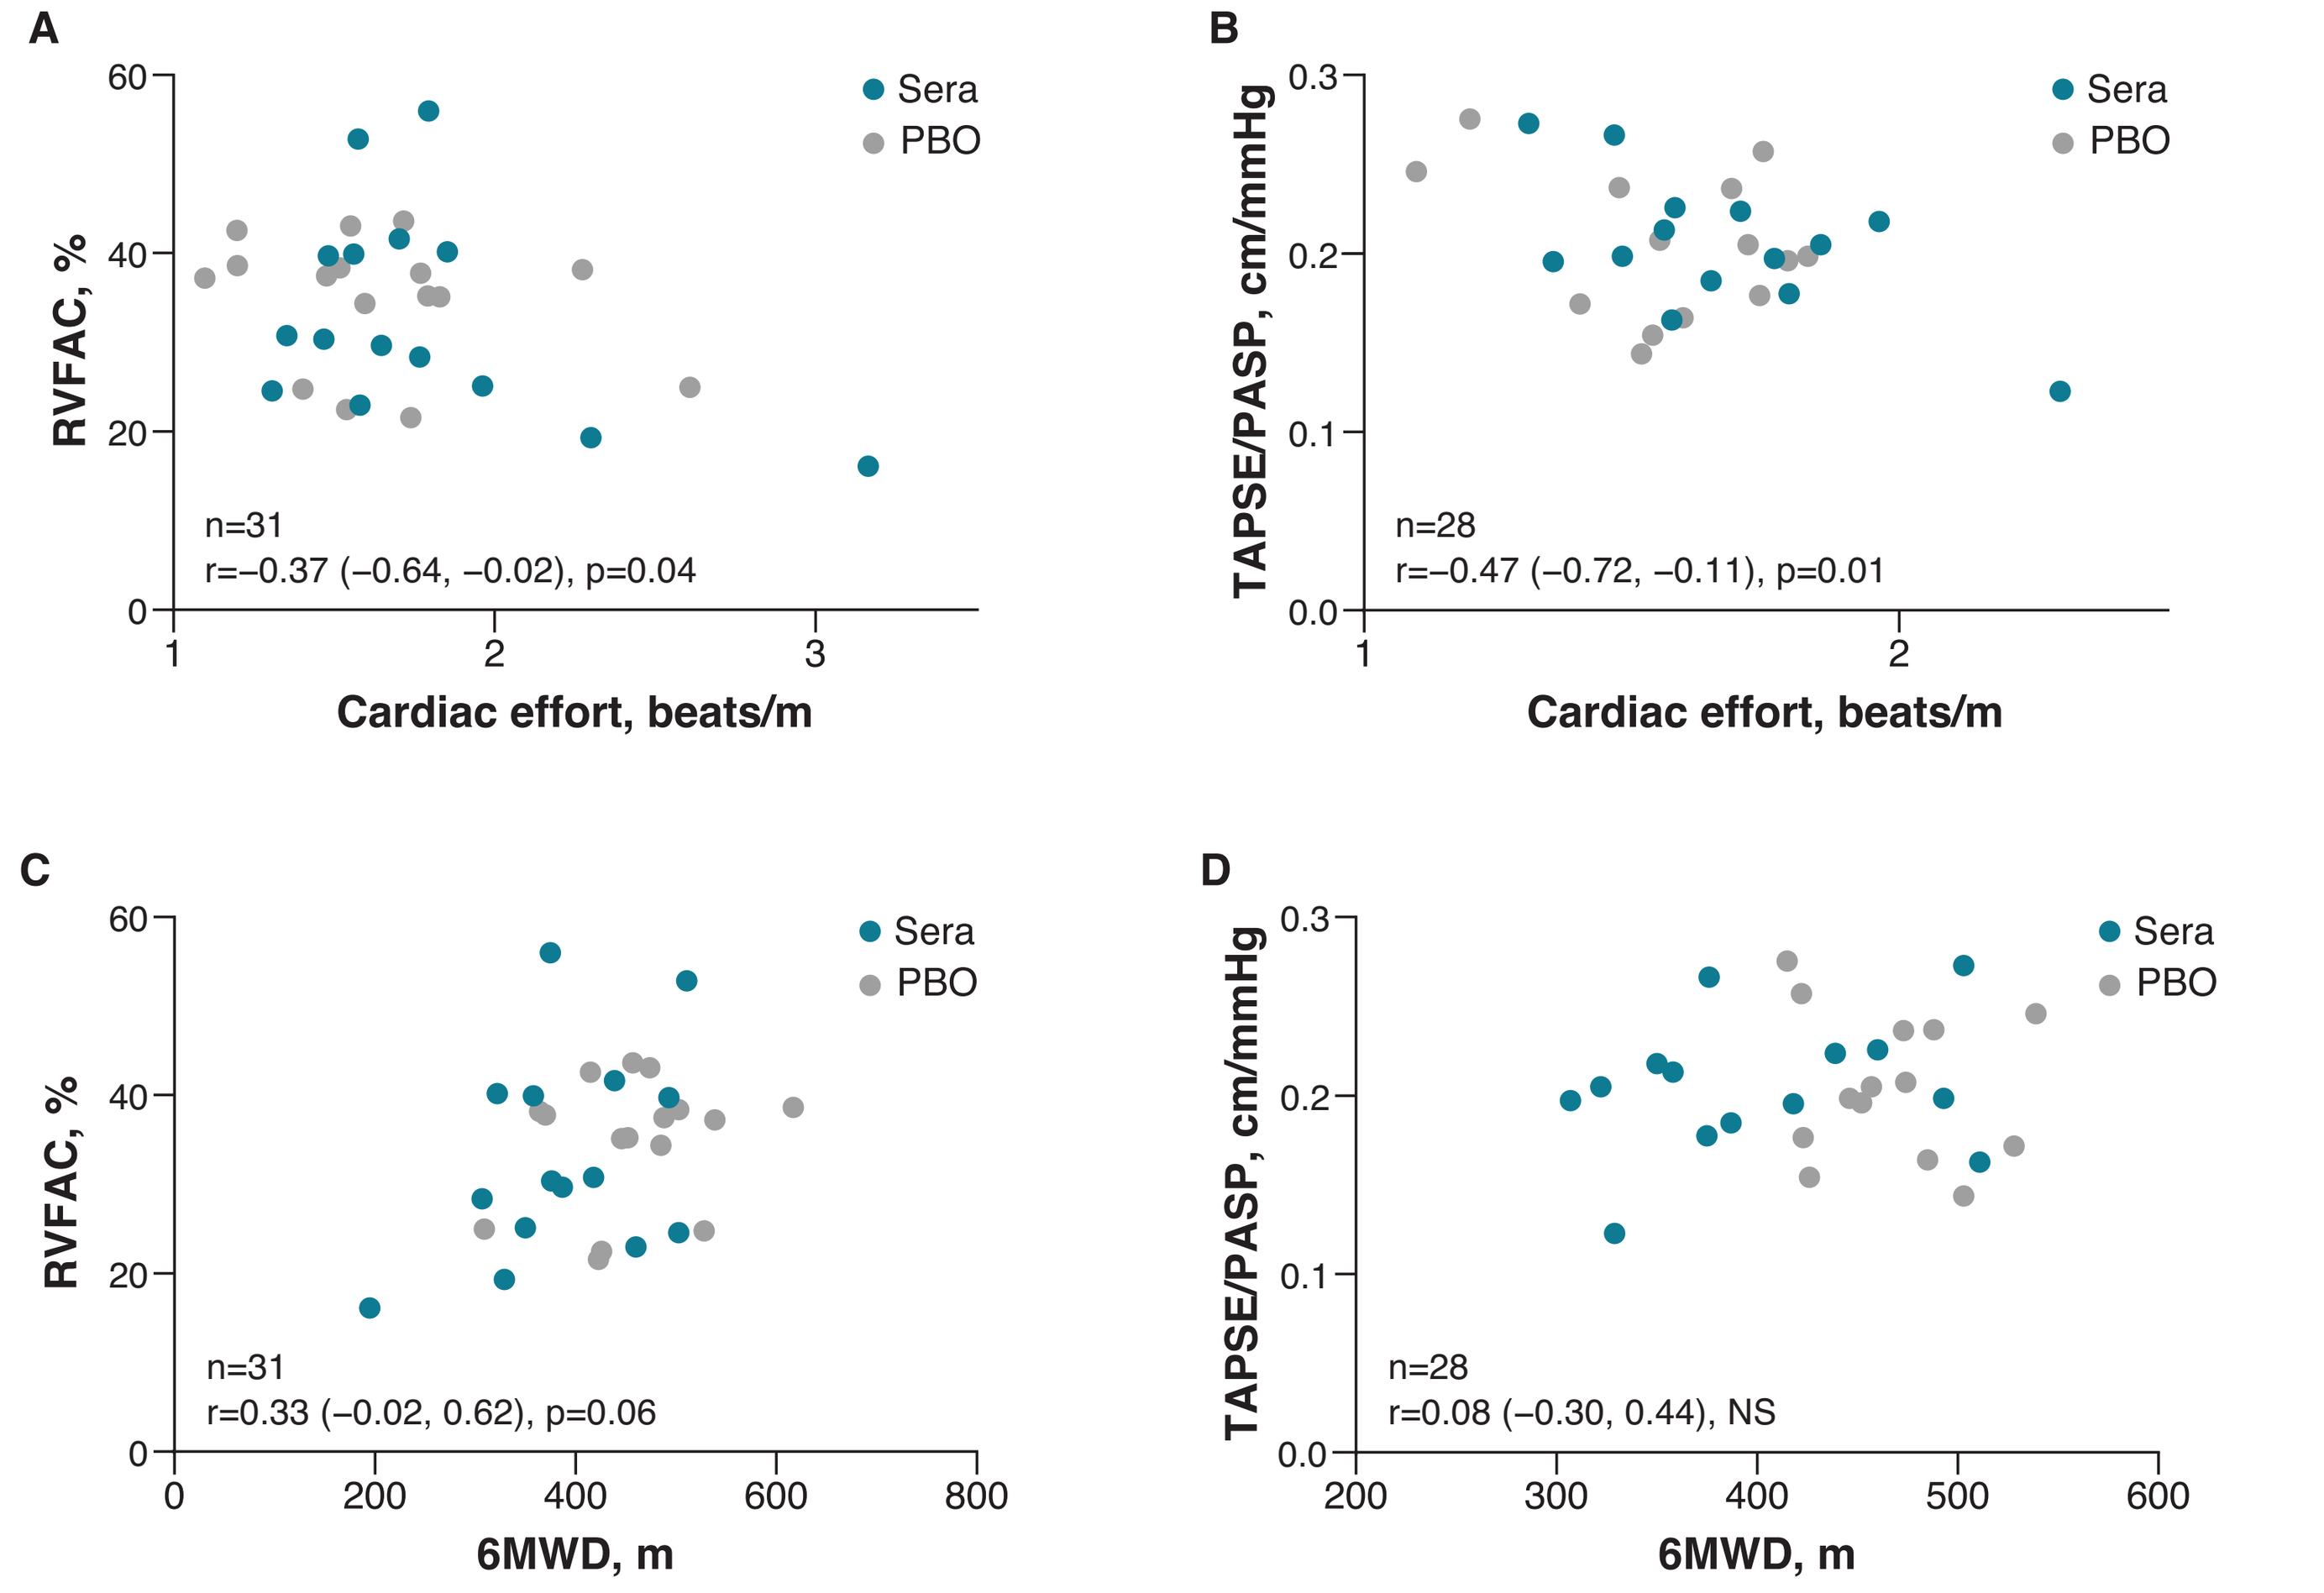
A
RVFAC, %
n=31
r=−0.37 (−0.64, −0.02), p=0.04
Cardiac effort, beats/m
B
TAPSE/PASP, cm/mmHg
n=28
r=−0.47 (−0.72, −0.11), p=0.01
Cardiac effort, beats/m
C
RVFAC, %
n=31
r=0.33 (−0.02, 0.62), p=0.06
6MWD, m
D
TAPSE/PASP, cm/mmHg
n=28
r=0.08 (−0.30, 0.44), NS
6MWD, m
Sera
PBO

Figure 4. A, B. Cardiac effort (CE) correlated with right ventricular fractional area change (RVFAC) and tricuspid annular plane systolic excursion (TAPSE)/pulmonary artery systolic pressure (PASP; an approximation of RV-PA coupling). **C, D.** 6MWD correlated with RVFAC but not with TAPSE/PASP. Right atrial area not shown; 6MWD correlated slightly better than CE with that measure. Pearson (parametric) correlations are shown. Repeated measures are included for some patients. 6MWD, six-minute walk distance; CE, cardiac effort; NS, not significant; PA, pulmonary artery; PBO, placebo; RV, right ventricle; Sera, seralutinib.

CONCLUSIONS

Cardiac effort

- Had less variability than 6MWD in placebo-treated patients
- Uncovered physiologic changes not apparent with 6MWD alone
- May reflect a treatment effect and may be used as a clinical endpoint to demonstrate a treatment effect
- Was associated with changes in REVEAL Lite 2 risk score
- Had better correlations with NT-proBNP and echocardiographic parameters than 6MWD

Limitations

- This was a substudy with very small patient numbers, ~20% heart rate data loss, and an imbalance in Baseline characteristics
- Data loss had two key sources: COVID-19 related issues (no hands-on training for a new technology, coordinator turnover), and unreliable heart rate data transmission during 6MWT
- Alternative devices that address these limitations can be explored in future studies

References:
1. Humbert M, et al. *Eur Heart J* 2022; 43(38):3618-3731
2. Halliday SJ, et al. *Respir Med* 2020; 165:105933
3. Lachant DJ, et al. *Pulm Circ* 2024; 14(2):e12355
4. Lachant DJ, et al. *Pulm Circ* 2020; 10(4):2045894020972572
5. Lachant DJ, et al. *Ann Am Thorac Soc* 2020; 17(3):372-375
6. Frantz RP, et al. *Lancet Resp Med* Published online May 2, 2024. doi: https://doi.org/10.1016/S2213-2600(24)00072-9

Acknowledgements: The authors would like to thank the study investigators, study coordinators, and especially the patients and families who participated in this study.

Research supported by: Gossamer Bio, Inc.

bitly