1045

Phase 2 Clinical Study to Evaluate the Efficacy and Safety of Inhaled Seralutinib for the Treatment of World Health Organization Group 1 Pulmonary Arterial Hypertension

Robert P. Frantz[1], Luke S. Howard[2], Vallerie V. McLaughlin[3], Olivier Sitbon[4], Roham T. Zamanian[5], Raymond L. Benza[6], Kelly Chin[7], Richard Channick[8], Matt Cravets[9], Jean-Marie Bruey[9], Robert Roscigno[9], David Mottola[9], Lawrence S. Zisman[9], Hossein-Ardeschir Ghofrani[10]

[1]Mayo Clinic, Rochester, MN, USA; [2]Imperial College Healthcare NHS, London, UK; [3]University of Michigan, Ann Arbor MI, USA; [4]Université Paris-Saclay, Le Kremlin-Bicêtre, France; [5]Stanford University, Menlo Park, CA, USA; [6]Ohio State University, Columbus, OH, USA; [7]UT Southwestern Medical Center, Dallas, TX, USA; [8]UCLA Medical Center, Los Angeles, CA, USA; [9]Gossamer Bio, Inc., San Diego, CA, USA; [10]Justus-Liebig University Giessen, Giessen, Germany

BACKGROUND

- Abnormal signaling of PDGFRα/β, CSF1R, and c-KIT, and BMPR2 deficiency, drive cellular overgrowth in the lung vasculature and play key roles in PAH progression[1,2,3] (**Figure 1**)
- Seralutinib is a small-molecule kinase inhibitor that targets these pathways and modulates BMPR2 (**Figure 1**)
- Seralutinib is administered by dry power inhalation and maximizes the therapeutic index by directly targeting diseased pulmonary arterioles, minimizing systemic exposure, and decreasing the potential off-target adverse effects

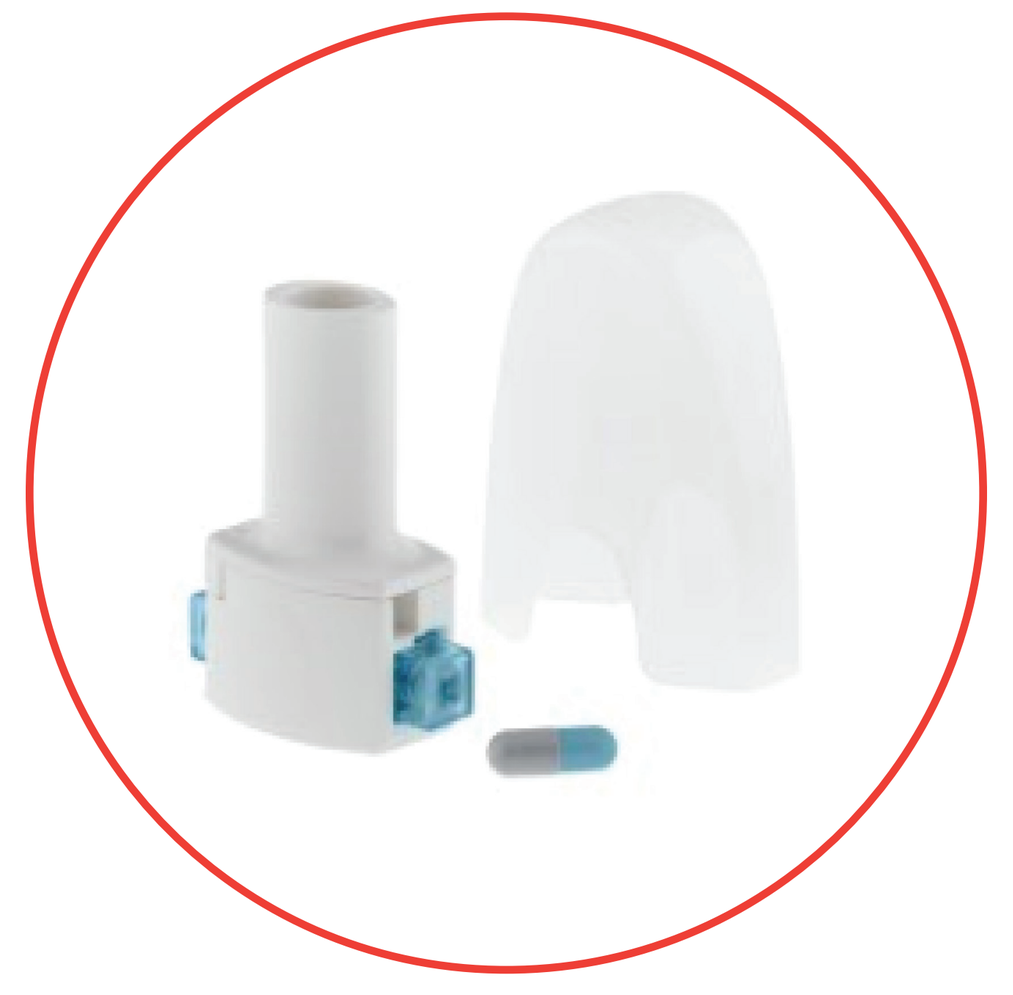

Dry Powder Inhaler

Figure 1. Mode of action of seralutinib in reversing pathologic remodeling in PAH

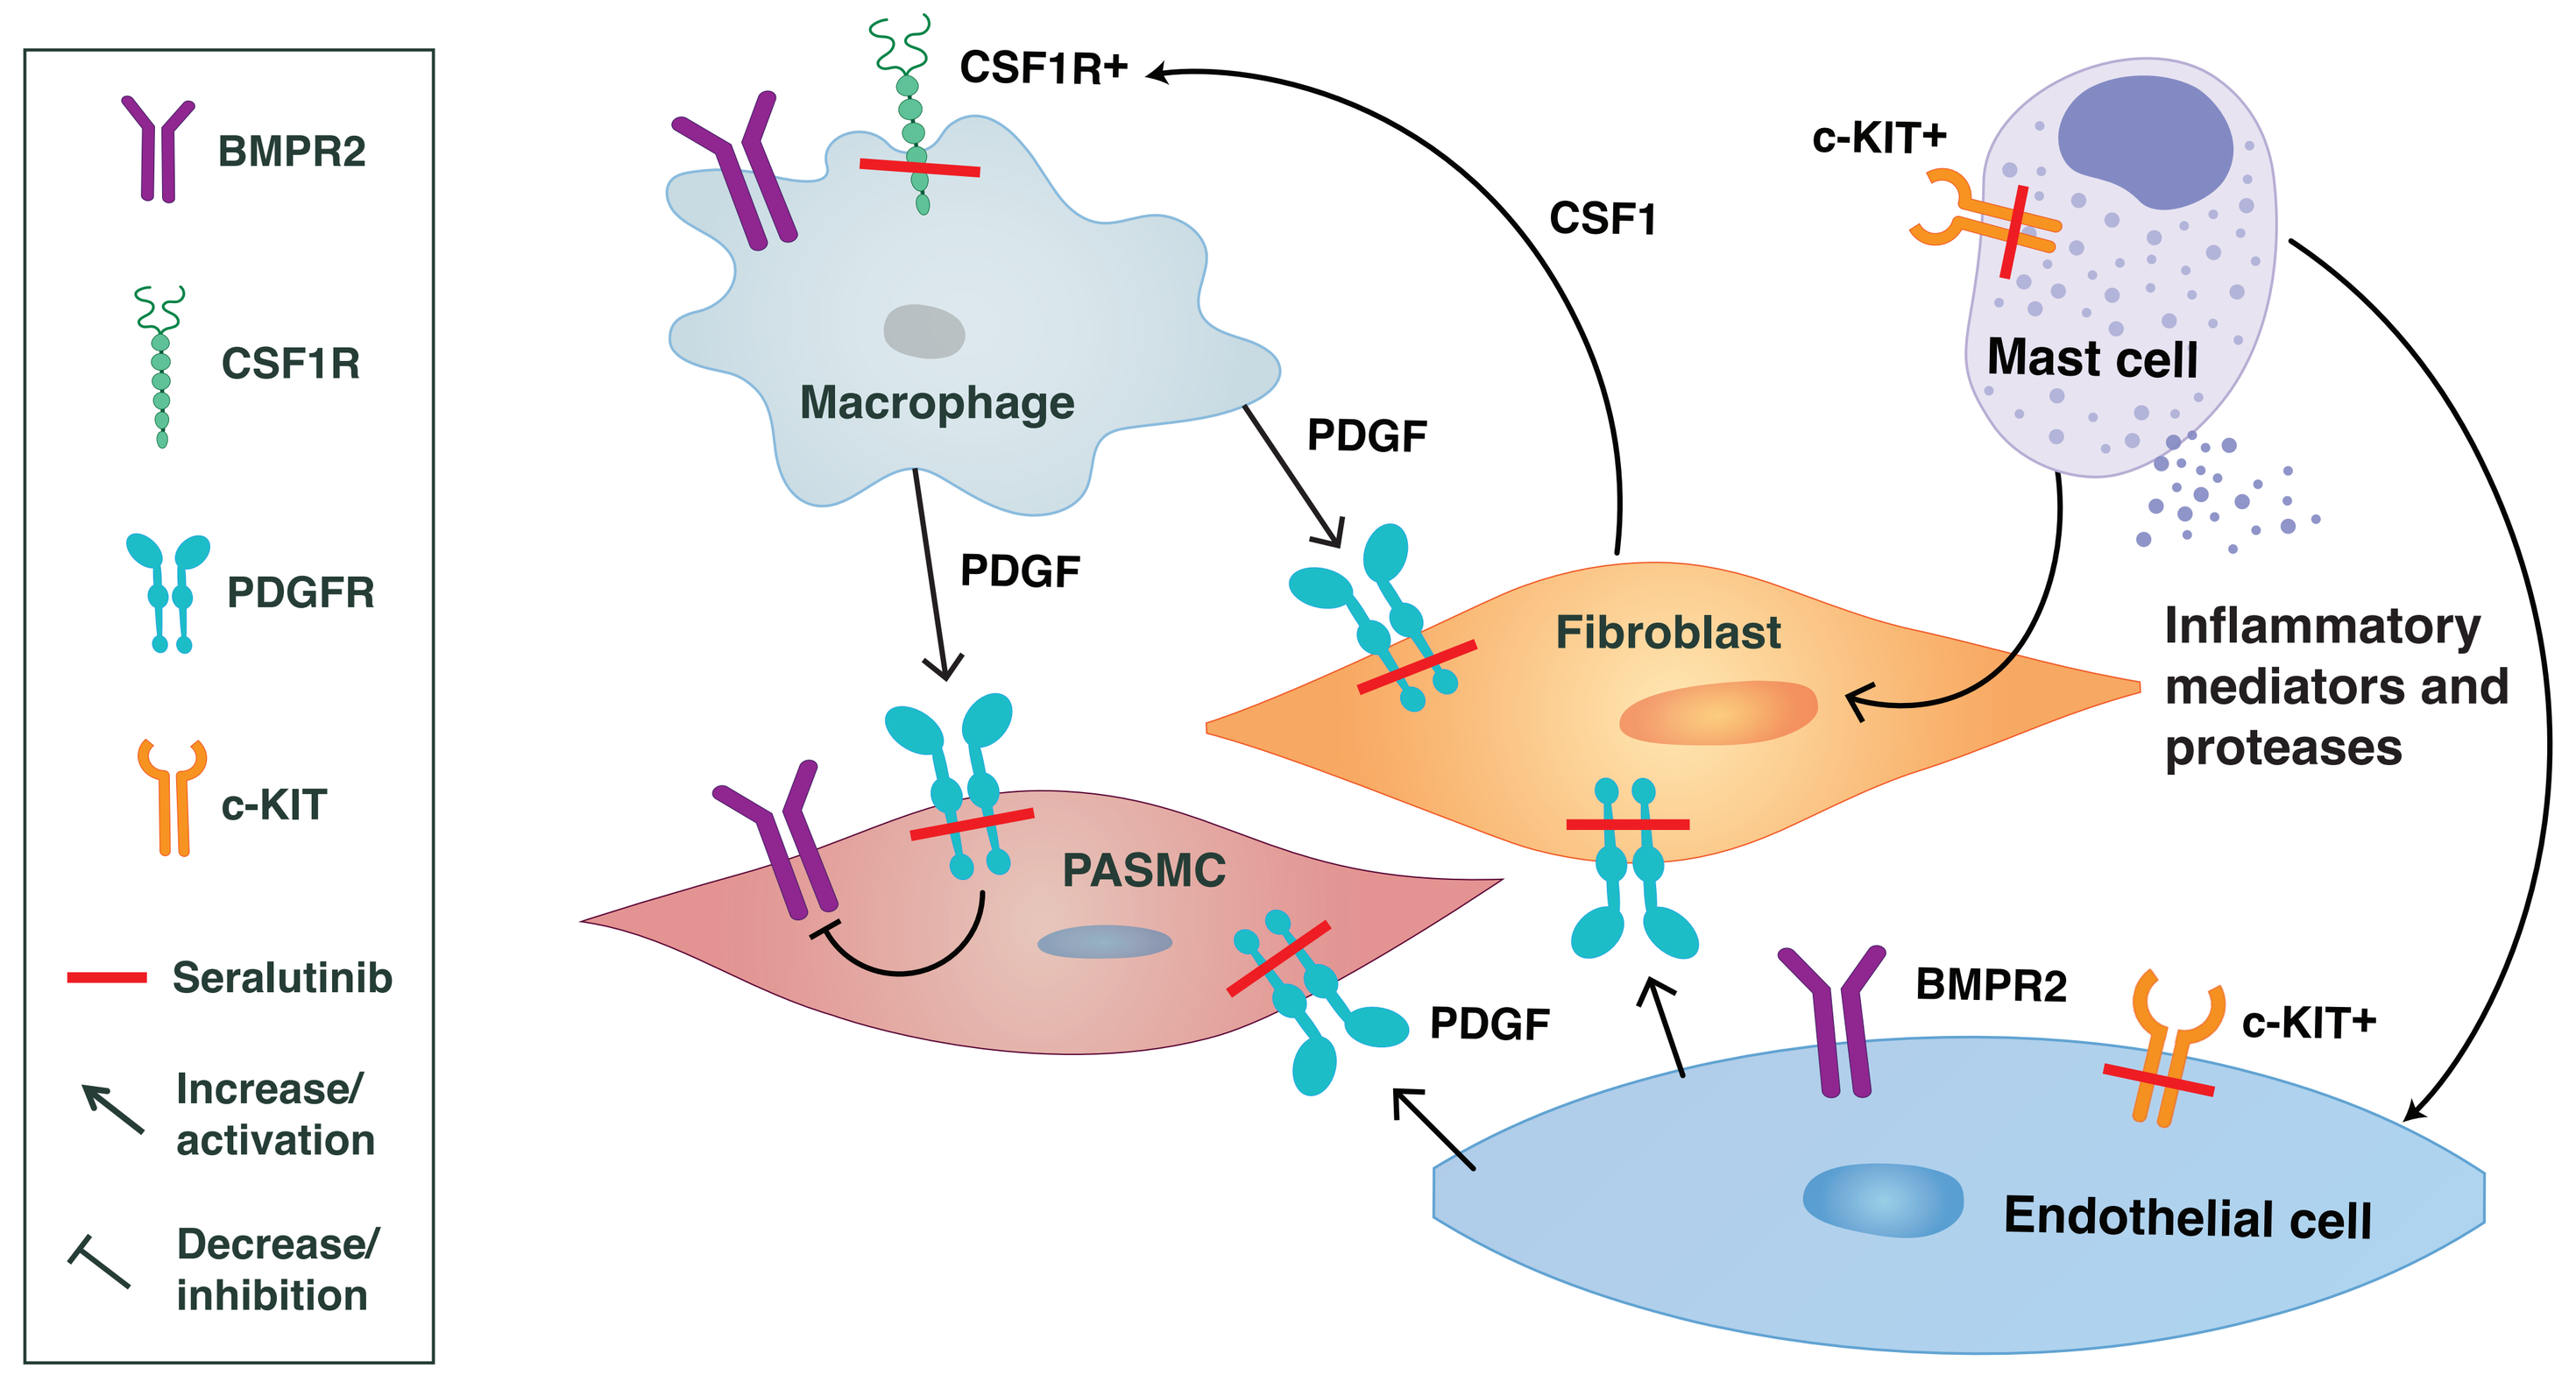

BMPR2
CSF1R
PDGFR
c-KIT
Seralutinib
Increase/ activation
Decrease/ inhibition
CSF1R+
CSF1
Macrophage
PDGF
PDGF
c-KIT+
Mast cell
Fibroblast
Inflammatory mediators and proteases
PASMC
PDGF
BMPR2
c-KIT+
Endothelial cell

- Seralutinib was evaluated in two preclinical animal models, monocrotaline/ pneumonectomy[4] and sugen 5416/hypoxia[5]
 - these models replicate key features of human PAH, including the development of neointimal proliferative lesions in the pulmonary arterioles and severe pulmonary hypertension
 - seralutinib reversed pulmonary vascular remodeling, improved hemodynamic parameters, reduced N-terminal pro-B-type natriuretic peptide (NT-proBNP), and increased lung BMPR2 expression vs controls[5]
- **Phase 1 studies** in healthy and PAH participants showed that seralutinib at doses up to 90 mg twice daily was well tolerated and achieved higher concentrations in lung tissue vs plasma[6]

TORREY STUDY

- The TORREY study is a randomized, double-blind, placebo-controlled trial (NCT04456998) designed to examine the efficacy and safety of inhaled seralutinib in subjects with PAH over a 24-week course of treatment

SELECTED INCLUSION CRITERIA

- Diagnosis of symptomatic PAH
- 6MWD ≥ 150 meters and ≤ 550 meters
- WHO FC II or III
- Treatment with standard of care PAH background therapies, including PGIs
- RHC data consistent with the diagnosis of PAH and PVR ≥ 400 dyne·s/cm5

SELECTED EXCLUSION CRITERIA

- Evidence of chronic thromboembolic disease or acute pulmonary embolism
- WHO Pulmonary Hypertension Group 2–5
- HIV-associated PAH
- History of left-sided heart disease and/or clinically significant cardiac disease
- Inhaled prostanoids
- Use of anticoagulants

	PRIMARY	SECONDARY
OBJECTIVES	Determine effect of seralutinib on pulmonary hemodynamics	Determine effect of seralutinib on exercise capacity
ENDPOINTS	Change in pulmonary vascular resistance from Baseline to Week 24	Change in six-minute walk distance from Baseline to Week 24 (Δ6MWD)

Study Schema: TORREY and Open-Label Extension Study

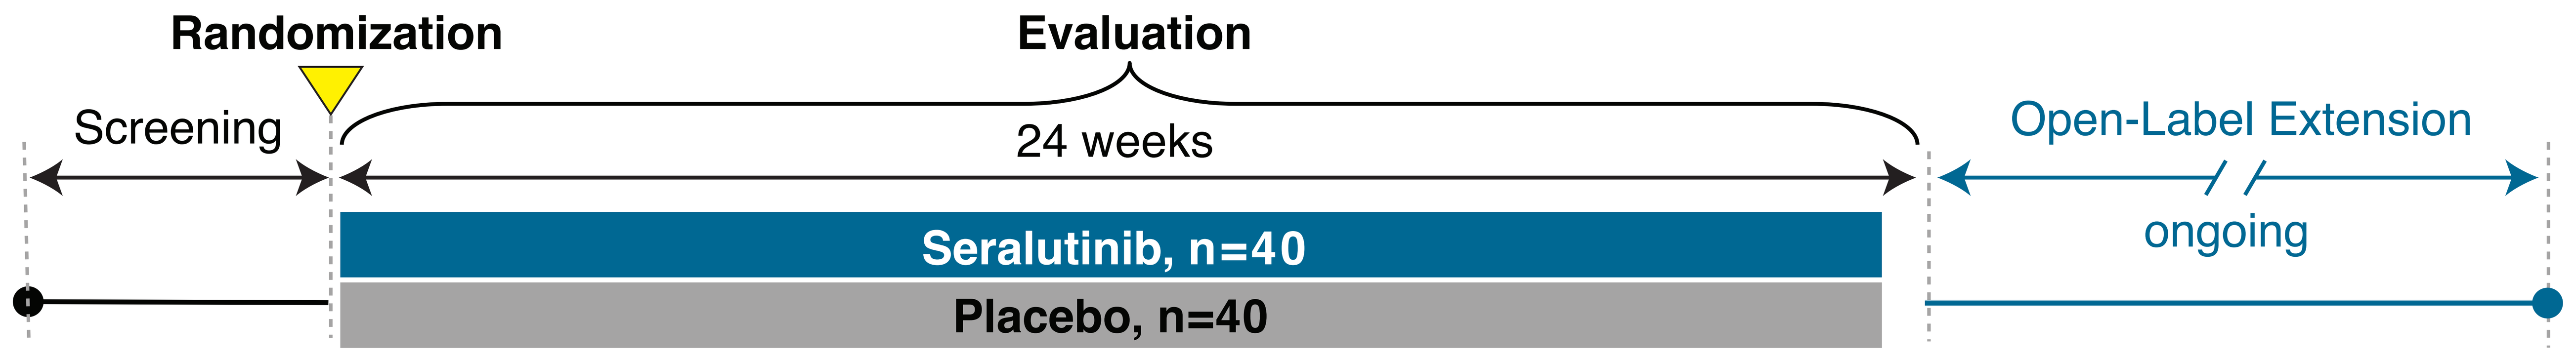

Randomization
Evaluation
Screening
24 weeks
Open-Label Extension
ongoing
Seralutinib, n=40
Placebo, n=40

Open-Label Extension (OLE) Study of Seralutinib

- Subjects may be eligible to participate in an OLE study (NCT04816604) provided they have completed study treatment and all associated assessments of TORREY or another seralutinib study
- Objectives are to evaluate the long-term safety, tolerability, and efficacy of inhaled seralutinib in subjects with PAH
- Subjects will receive seralutinib for up to 18 months
- Safety data will be collected periodically through week 72; right heart catheterization will be performed at week 48 to evaluate ongoing efficacy on pulmonary vascular resistance

TORREY SUBSTUDIES

Two substudies will explore novel endpoints relevant to the underlying pathophysiology of PAH

- **Functional Respiratory Imaging Substudy**
 This substudy will examine the effect of seralutinib on pulmonary vascular remodeling by quantifying changes in pulmonary arterial blood volume
- **Heart Rate Expenditure Substudy**
 This separate substudy will assess the effect of seralutinib on cardiac effort during the 6-minute walk test

SUMMARY

- **Seralutinib is a unique, inhaled, small-molecule kinase inhibitor that targets PDGFRα/β, CSF1R, and c-KIT, and modulates BMPR2**
- **The inhaled route of administration for seralutinib targets the diseased pulmonary arterioles at doses predicted to be locally effective while limiting systemic exposure which may reduce the risk of adverse events**
- **A phase 2 trial (TORREY; NCT04456998) in subjects with WHO Group 1 PAH is ongoing**
- **Eligible subjects may participate in a 72-week open-label extension study**

REFERENCES

1. Yamamura et al. *FASEB J* 2019; 33:7363-74.
2. Chen et al. *BMC Genomics* 2016; 17:781.
3. Perros et al. *Am J Respir Crit Care Med* 2008; 178:81-8.
4. Sitapara et al. *Circulation* 2019, Vol 140, A12947.
5. Galkin et al. *Circulation* 2019;140: A11102.
6. Li et al. *Am J Resp Crit Care Med* 2020; 201:A2907.

Presented at the PHA 2022 International PH Conference and Scientific Sessions, June 10-12, Atlanta, GA